Poster #: 14934

Six-Minute Walk Test (6MWT) Performance in Adult Patients Receiving Amikacin Liposome Inhalation Suspension (ALIS) for Treatment-Refractory Nontuberculous Mycobacterial (NTM) Lung Disease Caused by *Mycobacterium avium* Complex (MAC)

KL Winthrop,[1] G Eagle,[2] R Thomson,[3] TR Aksamit,[4] N Hasegawa,[5] K Morimoto,[6] D Addrizzo-Harris,[7] AE O'Donnell,[8] T Marras,[9] P Flume,[10] MR Loebinger,[11] L Morgan,[12] L Codecasa,[13] AT Hill,[14] SJ Ruoss,[15] J-J Yim,[16] FC Ringshausen,[17] S Field,[18] J Nezamis,[2] DE Griffith[19]

[1]OHSU-PSU School of Public Health, Portland, OR; [2]Insmed Incorporated, Bridgewater, NJ; [3]University of Queensland, Gallipoli Medical Research Institute, Brisbane, QLD, Australia; [4]Pulmonary Disease and Critical Care Medicine, Mayo Clinic, Rochester, MN; [5]Keio University Hospital, Tokyo, Japan; [6]Japan Anti-Tuberculosis Association Fukujuji Hospital, Tokyo, Japan; [7]Division of Pulmonary, Critical Care and Sleep Medicine, New York University School of Medicine, New York, NY; [8]Georgetown University Hospital, Washington, DC; [9]Department of Medicine, University of Toronto, and Toronto Western Hospital, University Health Network, Toronto, ON, Canada; [10]Medical University of South Carolina, Charleston, SC; [11]Host Defense Unit, Royal Brompton Hospital, and Imperial College, London, UK; [12]Concord Hospital, Concord, NSW, Australia; [13]TB Reference Centre, Villa Marelli Institute/Niguarda Hospital, Milan, Italy; [14]Department of Respiratory Medicine, Royal Infirmary and University of Edinburgh, Edinburgh, UK; [15]Division of Pulmonary and Critical Care, Department of Medicine, Stanford University School of Medicine, Stanford, CA; [16]Division of Pulmonary and Critical Care Medicine, Department of Internal Medicine, Seoul National University College of Medicine, Seoul, South Korea; [17]Department of Respiratory Medicine, Hannover Medical School, and German Center for Lung Research (DZL), Hannover, Germany; [18]Cumming School of Medicine, University of Calgary, Calgary, AB, Canada; [19]University of Texas Health Science Center, Tyler, TX

BACKGROUND

- Nontuberculous mycobacterial (NTM) lung disease is a chronic debilitating condition that can significantly impact morbidity and mortality.[1] *Mycobacterium avium* complex (MAC) accounts for 80% to 85% of cases in the USA.[2]
- Options are limited for patients with MAC lung disease in whom initial guideline-based therapy (GBT) has failed.[3]
- Amikacin, a potent antimycobacterial agent, is generally reserved for patients with fibrocavitary MAC disease or severe nodular/bronchiectatic disease due to its renal and auditory toxicity when administered parenterally.[3,4]
- Amikacin liposome inhalation suspension (ALIS) is amikacin sulfate (623 mg per vial amikacin base; to deliver 590 mg amikacin base to the nebulizer) encapsulated in liposomes for inhalational delivery. ALIS increases amikacin uptake into alveolar macrophages, a refuge for NTM organisms, and was designed to limit systemic amikacin exposure.[5,6]
- CONVERT is a randomized, open-label, phase 3 study that assessed the efficacy of adding ALIS to GBT in adult patients with treatment-refractory MAC lung disease
- MAC lung disease can adversely impact functional capacity.[7] The 6-minute walk test (6MWT) has been used to assess functional capacity across various cardiorespiratory diseases.[8]
- Although the 6MWT has not been validated in MAC lung disease, a phase 2 study of ALIS in patients with NTM lung disease showed improvement in this test, particularly in patients who achieved sputum culture conversion.[9]

OBJECTIVE

- Assess the effects of 6 months of treatment with ALIS + GBT on 6MWT performance in the CONVERT study

METHODS

Key inclusion criteria

- Age ≥ 18 years with MAC lung disease
 - At least 2 MAC-positive sputum or bronchoscopy cultures within 6 months, despite ≥ 6 months of GBT either ongoing or stopped within 12 months before screening
 - Characteristic lung findings (eg, nodular infiltrate, cavity) on a chest radiograph or chest computed tomography

Key exclusions

- Cystic fibrosis, active pulmonary tuberculosis, immunodeficiency syndromes, MAC isolates with amikacin minimum inhibitory concentration > 64 μg/mL, active malignancies within 1 year

Treatment and assessments

- Patients were randomly assigned (2:1) to receive ALIS + GBT or GBT alone (**Figure 1**). ALIS was inhaled once daily by eFlow nebulizer.
- Patients with culture conversion (3 consecutive monthly MAC-negative sputum cultures) by Month 6 received 12 months of further treatment after the first conversion-defining negative culture
- Patients from either arm who did not convert by Month 6 had the option to enroll in an extension study (INS-312) and receive continued or add-on ALIS
- Monthly clinic visits included sputum collection in duplicate or (preferably) triplicate, assessment of adverse events, physical examinations, and standard laboratory tests
- The 6MWT was administered at Baseline and Months 4 and 6; audiology tests were performed at Baseline and Months 3 and 6

Figure 1. CONVERT: Randomized, Open-Label Phase 3 Study

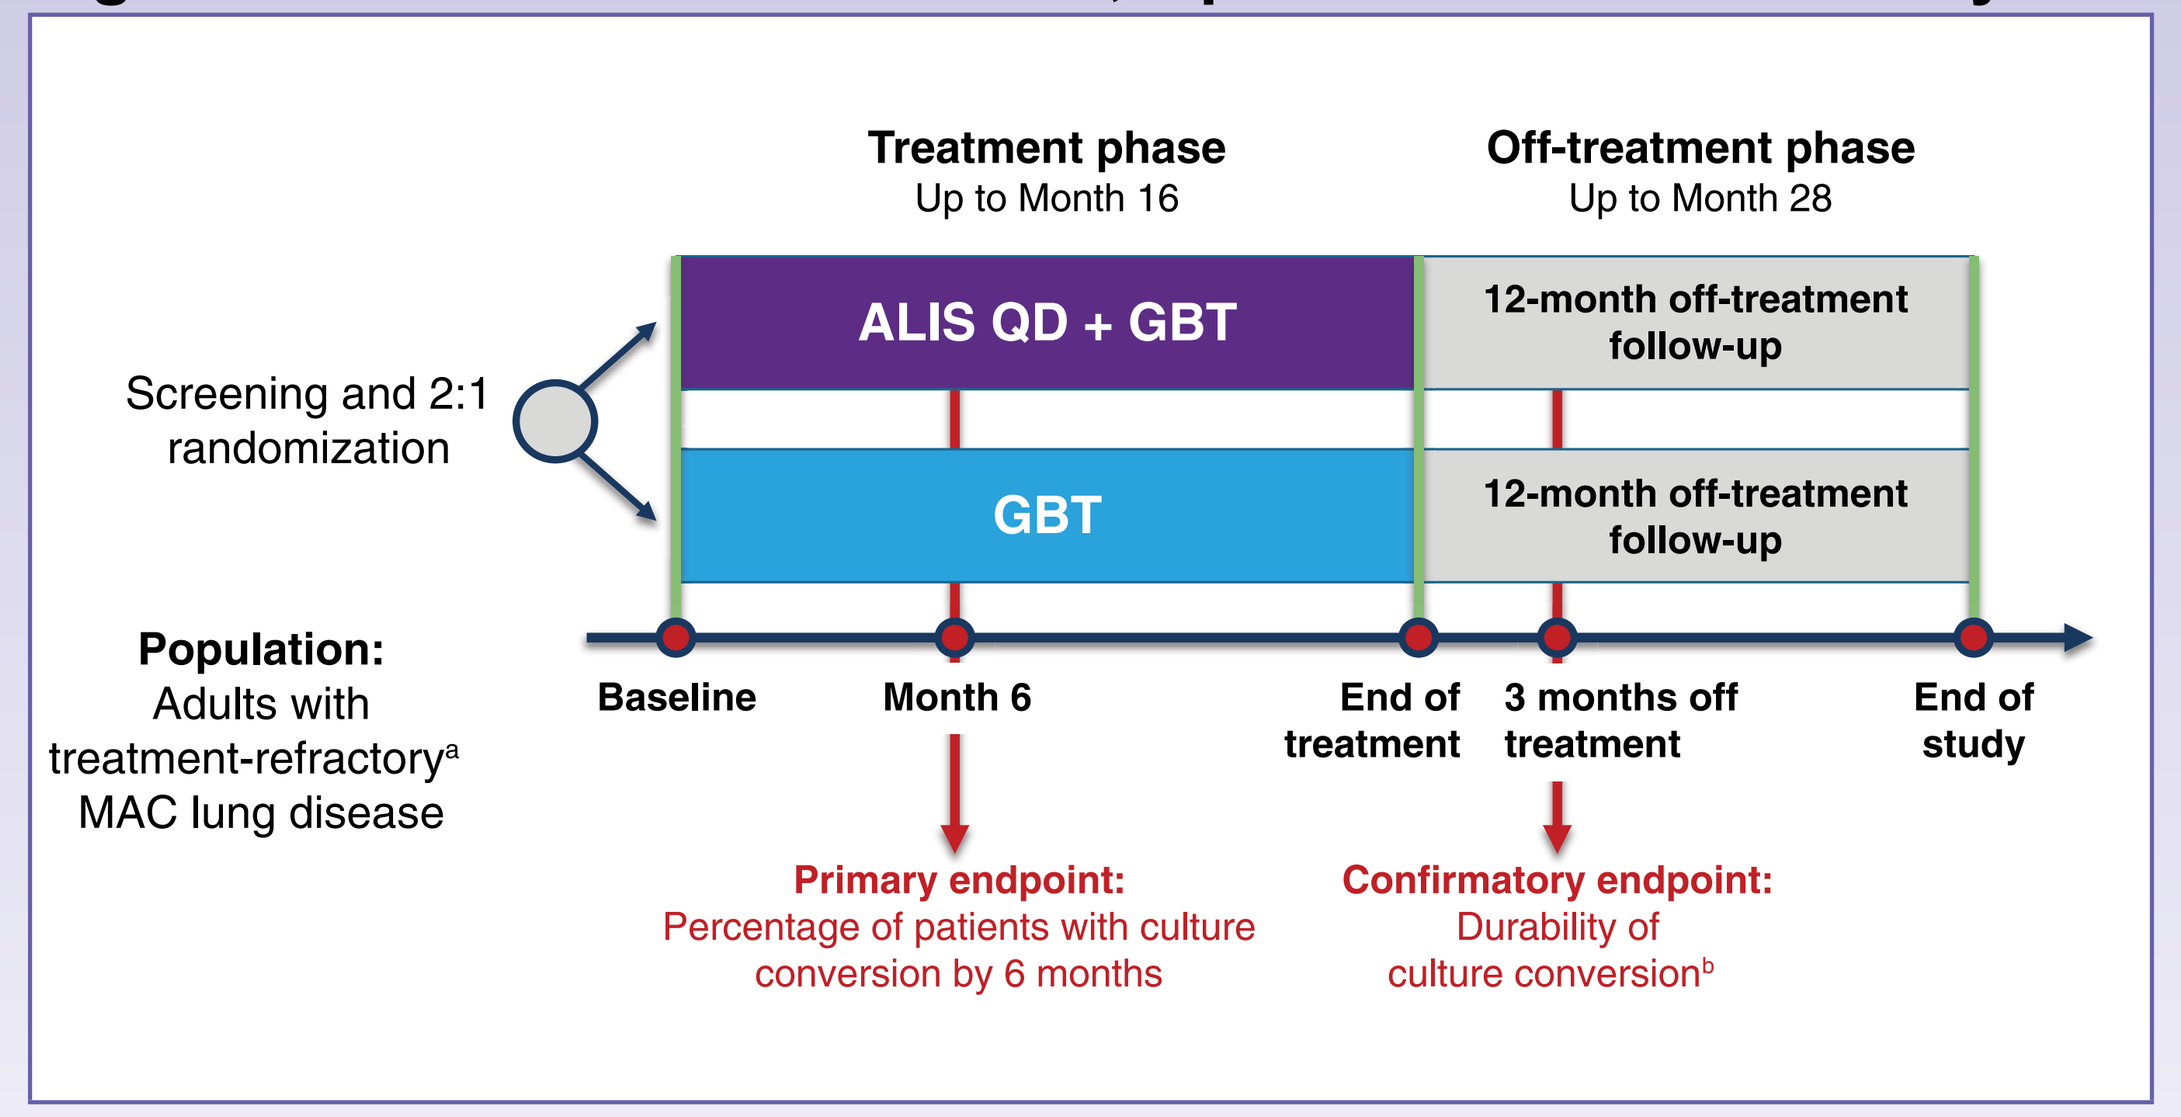
Treatment phase
Up to Month 16
Off-treatment phase
Up to Month 28
Screening and 2:1 randomization
ALIS QD + GBT
12-month off-treatment follow-up
GBT
12-month off-treatment follow-up
Population:
Adults with treatment-refractory[a] MAC lung disease
Baseline
Month 6
End of treatment
3 months off treatment
End of study
Primary endpoint:
Percentage of patients with culture conversion by 6 months
Confirmatory endpoint:
Durability of culture conversion[b]

[a] At least 6 months of treatment with sputum cultures persistently positive for MAC.
[b] Final analysis at completion of study is durability of culture conversion 3 months off all MAC treatment for patients who complete 12 months of treatment from the first negative culture that defined conversion.
ALIS, amikacin liposome inhalation suspension; GBT, guideline-based therapy; MAC, *Mycobacterium avium* complex; QD, once daily.

Endpoints	
Primary endpoint	• Sputum culture conversion by Month 6 following addition of ALIS (590 mg) + GBT compared with GBT alone[10] – Defined as 3 consecutive monthly MAC-negative sputum cultures – To meet this endpoint, initial detection of a negative sputum culture had to occur by Month 4
Key secondary endpoint	• **Change in distance achieved in the 6MWT from Baseline to Month 6 following addition of ALIS (590 mg) + GBT compared with GBT alone**
Prespecified exploratory endpoints	• Change in 6MWT distance from Baseline to Month 6 by sputum conversion status, overall and by treatment group

RESULTS

Patients

- 224 and 112 patients were assigned to ALIS + GBT and GBT alone, respectively
- At the time of data analysis, 19.6% of patients in the ALIS + GBT arm and 8.9% of patients in the GBT alone arm had withdrawn from the study (**Table 1**)
- The mean age was 64.7 years; most patients were female (69.3%) and white (69.9%) (**Table 2**)
- The most common concomitant lung diseases included bronchiectasis (62.5%), chronic obstructive pulmonary disease (COPD; 14.3%), or both (11.9%)

Table 1. Patient Disposition

	ALIS + GBT	GBT Alone	Total
Screened			492
Screen Failure			156
Intent to Treat population (ITT)[a]	224	112	336
Safety Population[b]	223	112	335
Study Withdrawal	44 (19.6%)	10 (8.9%)	54 (16.1%)

[a] All randomized patients
[b] Randomized patients who received at least 1 dose of ALIS (ALIS + GBT arm) or GBT (GBT-alone arm)
As of data cutoff on July 7, 2017.

Table 2. Patient Demographic and Disease Characteristics.

Parameter	ALIS + GBT N = 224	GBT Alone N = 112	Total N = 336
Age, mean years (SD)	64.6 (9.6)	64.9 (10.2)	64.7 (9.8)
Female, n (%)	165 (73.7)	68 (60.7)	233 (69.3)
Body mass index, mean kg/m² (SD)	21.4 (3.9)	21.0 (3.8)	NA
Race, n (%)			
White	158 (70.5)	77 (68.8)	235 (69.9)
Japanese	35 (15.6)	15 (13.4)	50 (14.9)
Other Asian	23 (10.3)	10 (8.9)	33 (9.8)
Other/not reported	5 (2.2)	7 (6.3)	12 (3.6)
Black/African American	3 (1.3)	3 (2.7)	6 (1.8)
Underlying lung disease, n (%)			
Bronchiectasis only	146 (65.2)	64 (57.1)	210 (62.5)
COPD only	29 (12.9)	19 (17.0)	48 (14.3)
COPD and bronchiectasis	22 (9.8)	18 (16.1)	40 (11.9)
Current smoker, n (%)	26 (11.6)	10 (8.9)	36 (10.7)

ALIS, amikacin liposome inhalation suspension; COPD, chronic obstructive pulmonary disease; GBT, guideline-based therapy; NA, not available.

Efficacy

- Culture conversion, the primary endpoint, was achieved by 65 of 224 patients (29.0%) receiving ALIS + GBT and 10 of 112 (8.9%) receiving GBT alone (P < 0.0001)[10]
- At Month 6, there was no statistically significant difference in change from Baseline in 6MWT between treatment groups (**Table 3**)
- In the overall population, patients who achieved culture conversion had greater improvement in 6MWT than patients without culture conversion (P = 0.011) (**Figure 2**)
 - 6MWT improvement was particularly evident in patients within the ALIS + GBT group who achieved culture conversion (P = 0.005)
 - The observed difference within the GBT-alone group did not achieve significance, which may be due to the small number of converters in that group

Table 3. 6MWT Response by Treatment Group.

Analysis Group (n[a])	Baseline Mean meters (SD)	Month 6 Mean meters (SD)	LS Mean Change, Baseline to Month 6 (95% CI)	P Value
ALIS + GBT (220)	425.7 (127.6)	425.2 (141.23)	−1.9 (−24.0, 20.2)	0.72
GBT Alone (111)	420.4 (126.7)	423.3 (132.73)	1.3 (−22.2, 24.9)	

[a] For Baseline, n is the number of patients with a Baseline score and at least one post-Baseline score. For Month 6, n is the number of patients with a Baseline score and a post-Baseline score at the summarized visit.
Baseline is defined as the last nonmissing value prior to first dose of study drug. A multiple imputation technique was used to account for missing data.
LS means were obtained from a MMRM model which included treatment, month, the treatment-by-month interaction, smoking status, and prior multidrug regimen as fixed factors, the baseline 6MWT distance as a covariate, and baseline 6MWT distance-by-month interaction.
6MWT, 6-minute walk test; ALIS, amikacin liposome inhalation suspension; GBT, guideline-based therapy; LS, least squares.

Figure 2. Difference in 6MWT Distance From Baseline Based on Sputum Conversion Status (Converters minus Non-Converters)

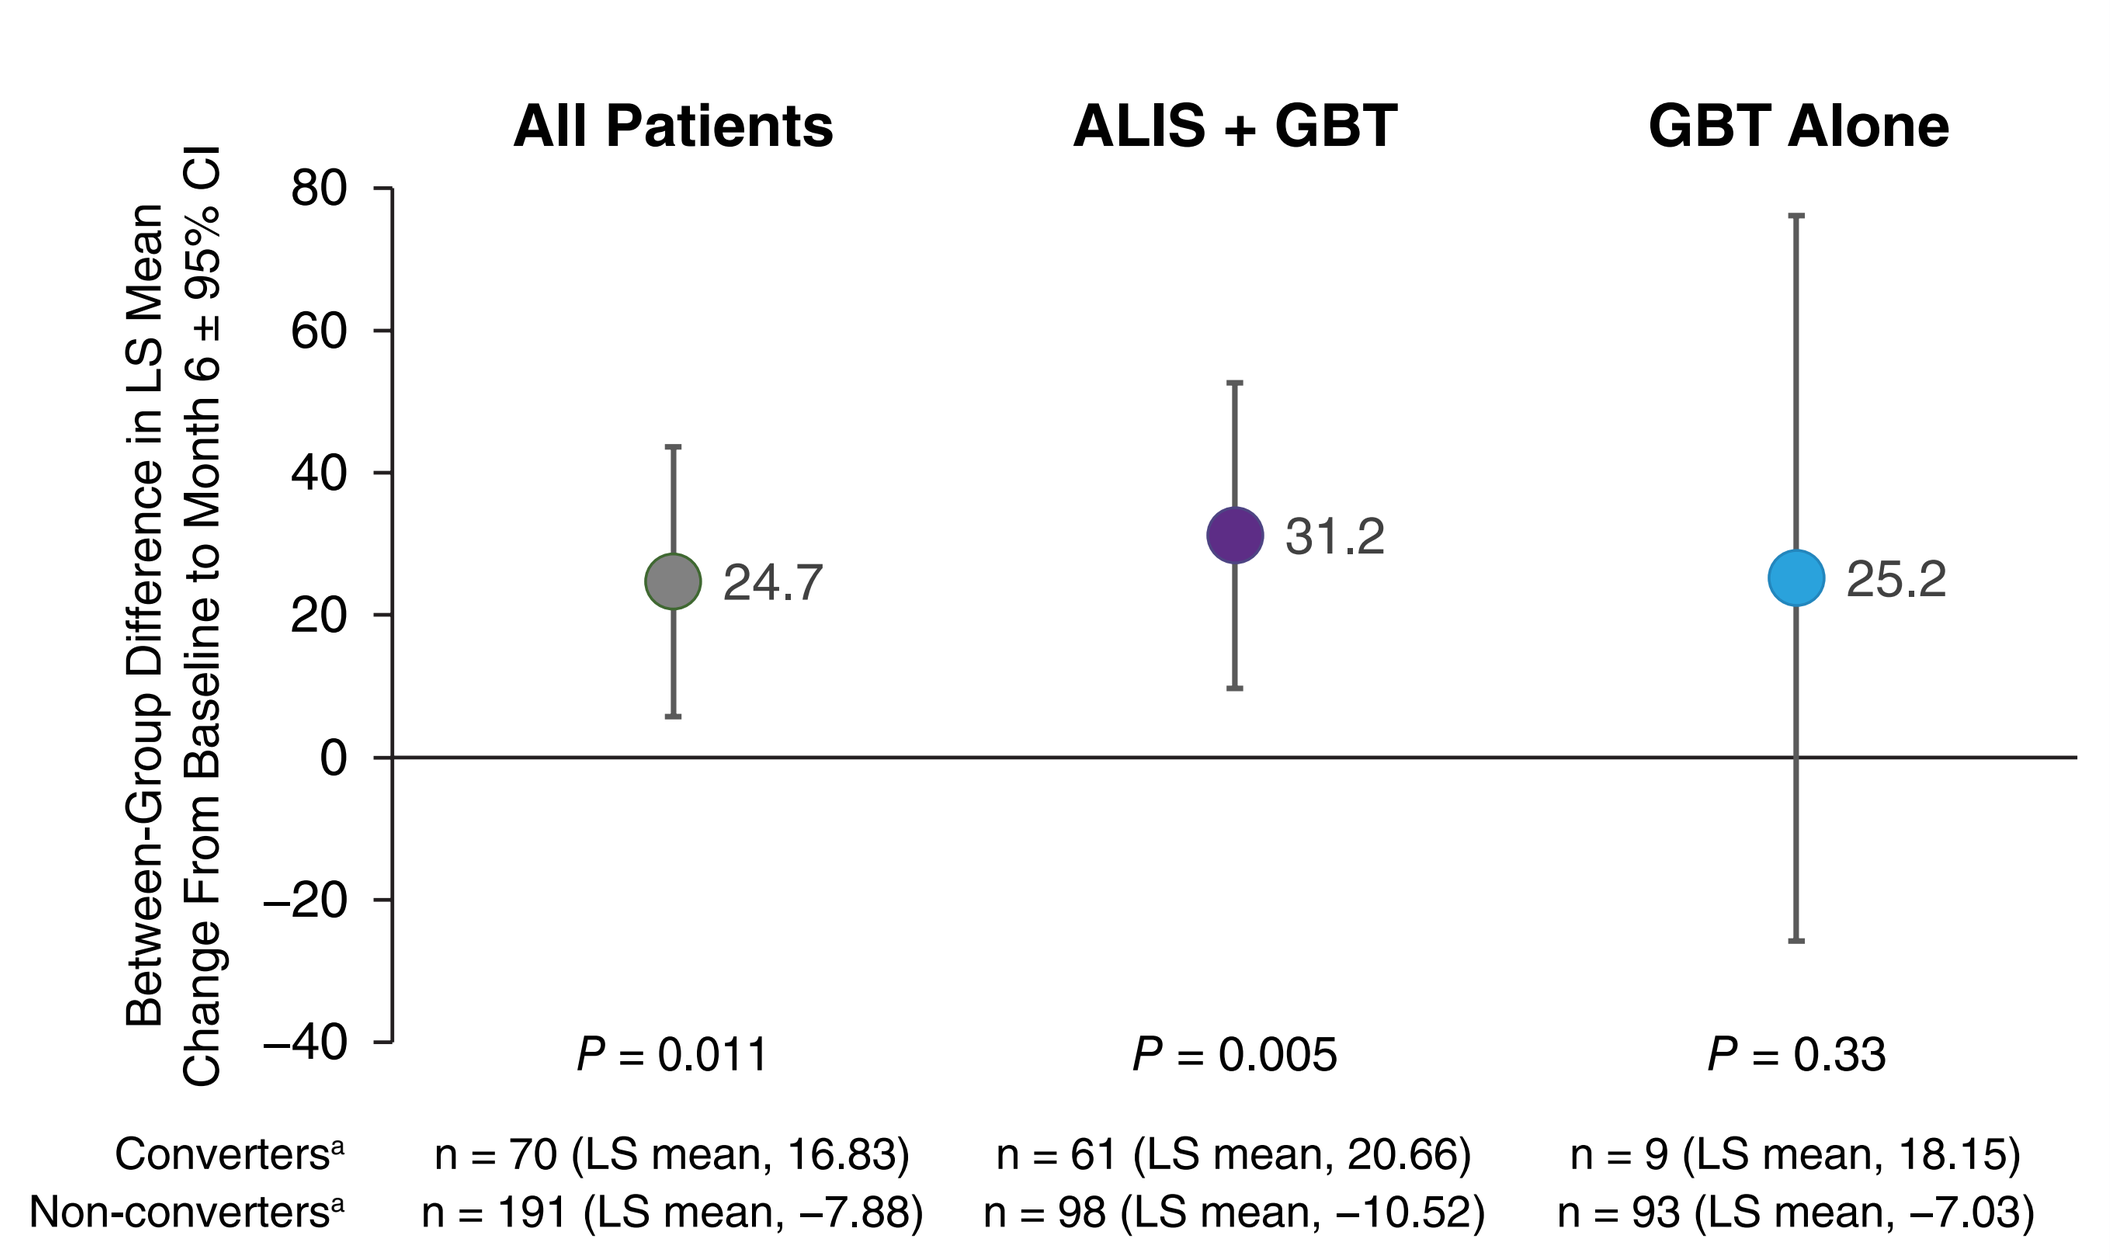
All Patients
ALIS + GBT
GBT Alone
Between-Group Difference in LS Mean Change From Baseline to Month 6 ± 95% CI
80
60
40
20
0
−20
−40
24.7
31.2
25.2
P = 0.011
P = 0.005
P = 0.33
Converters[a]
Non-converters[a]
n = 70 (LS mean, 16.83)
n = 191 (LS mean, −7.88)
n = 61 (LS mean, 20.66)
n = 98 (LS mean, −10.52)
n = 9 (LS mean, 18.15)
n = 93 (LS mean, −7.03)

[a] LS mean: change from Baseline in 6MWT distance (meters); n is the number of patients with a Baseline score and a post-Baseline score at the Month 6 visit.
ANCOVA model of change from Baseline to each visit in 6MWT distance, which includes converter status, the combination of smoking status and prior multidrug regimen as fixed factors, and the Baseline 6MWT distance as a covariate.
6MWT, 6-minute walk test; ALIS, amikacin liposome inhalation suspension; ANCOVA, analysis of covariance; GBT, guideline-based therapy; LS, least squares.

Safety

- Treatment-emergent adverse events (TEAE) were reported in 98.2% and 91.1% of patients in the ALIS + GBT and GBT-alone arms, respectively (**Table 4**)
 - Most events were of moderate severity in the ALIS + GBT arm and mild in the GBT-alone arm
- The most common TEAEs overall were respiratory in nature
 - Respiratory TEAEs were reported in 87.4% of patients receiving ALIS + GBT and 50.0% receiving GBT alone
 - TEAEs reported in ≥ 10% of patients in the ALIS + GBT arm included dysphonia, cough, hemoptysis, dyspnea, fatigue, diarrhea, nausea, and oropharyngeal pain
 - Most events of these types were initially reported in the first month of ALIS treatment, with declining incidence thereafter
- Serious TEAEs occurred in 20.2% and 17.9% of patients, respectively
- TEAE of Special Interest were reported in 54.7% of patients receiving ALIS + GBT and 30.4% receiving GBT alone (**Table 5**)
 - Most events of Special Interest were of mild to moderate severity
 - Tinnitus was higher in ALIS + GBT arm (7.6%) than in the GBR alone arm (0.9%). However, hearing loss was similar across arms (ALIS + GBT, 4.5%; GBT alone, 6.3%)

Table 4. Safety Summary

Preferred Term, n (%)	ALIS + GBT N = 223[a]	GBT Alone N = 112
Any TEAE	219 (98.2)	102 (91.1)
Any serious TEAE	45 (20.2)	20 (17.9)
TEAE leading to death	6 (2.7)	5 (4.5)
Serious TEAE occurring in ≥ 3% of patients in either arm		
Pneumonia	8 (3.6)	2 (1.8)
COPD exacerbation	7 (3.1)	1 (0.9)
Hemoptysis	6 (2.7)	5 (4.5)
TEAEs occurring in ≥ 10% of patients in either arm		
Dysphonia	102 (45.7)	1 (0.9)
Cough	83 (37.2)	17 (15.2)
Dyspnea	48 (21.5)	10 (8.9)
Hemoptysis	39 (17.5)	15 (13.4)
Fatigue	36 (16.1)	8 (7.1)
Diarrhea	28 (12.6)	5 (4.5)
Nausea	25 (11.2)	4 (3.6)
Oropharyngeal pain	24 (10.8)	2 (1.8)

[a] ALIS + GBT safety population (N = 223) differs from intention-to-treat population (N = 224) due to 1 patient who did not initiate study therapy.
ALIS, amikacin liposome inhalation suspension; COPD, chronic obstructive pulmonary disease; GBT, guideline-based therapy; TEAE, treatment-emergent adverse event.

Table 5. TEAEs of Special Interest

Preferred Term, n (%)	ALIS + GBT N = 223[a]	GBT Alone N = 112
Any TEAE of Special Interest[b]	122 (54.7)	34 (30.4)
TEAE of Special Interest Occurring in ≥ 3% of patients in either arm		
Dyspnea	48 (21.5)	10 (8.9)
Hemoptysis	39 (17.5)	15 (13.4)
Wheezing ± bronchospasm	20 (8.9)	3 (2.7)
COPD exacerbation	18 (8.1)	3 (2.7)
Tinnitus	17 (7.6)	1 (0.9)
Dizziness	14 (6.3)	3 (2.7)
Hearing loss[c]	10 (4.5)	7 (6.3)

[a] ALIS + GBT safety population (N = 223) differs from intention-to-treat population (N = 224) due to 1 patient who did not initiate study therapy.
[b] Defined as pre-specified events known to be attributed to parenteral amikacin and inhalation of antibiotics.
[c] Includes preferred terms Hypoacusis, Deafness neurosensory, and Deafness.
ALIS, amikacin liposome inhalation suspension; COPD, chronic obstructive pulmonary disease; GBT, guideline-based therapy; TEAE, treatment-emergent adverse event.

SUMMARY

- In the CONVERT study, the addition of ALIS to GBT improved culture conversion rates in patients with treatment-refractory MAC lung disease, compared with GBT alone.[10]
- The addition of ALIS to GBT was associated with higher rates of TEAEs, mostly respiratory in nature and predominantly mild or moderate in severity.
- While the difference in 6MWT distance was not statistically significant between treatment arms, a nominally significant increase in 6MWT distance was observed in patients with culture conversion vs a decrease in those without culture conversion (absolute difference, 24.71 meters).
- Improvements in 6MWT distance among converters suggest that culture conversion may benefit physical function.

REFERENCES

1. Adjemian J, et al. Am J Respir Crit Care Med 2012; 185:881-886.
2. Prevots DR, et al. Clin Chest Med 2015; 36:13-34.
3. Griffith DE, et al. Curr Opin Infect Dis 2012; 25:218-227.
4. Griffith DE, et al. Am J Respir Crit Care Med 2007; 175:367-416.
5. Zhang J, et al. Front Microbiol 2018; doi: 10.3389/fmicb.2018.00915
6. Rose SJ, et al. PLoS One 2014; 9:e108703.
7. Yeh JJ, et al. PLoS One 2014; 9:e99260.
8. Holland AE, et al. Eur Respir J 2014; 44:1428-1446.
9. Olivier KN, et al. Am J Respir Crit Care Med 2017; 195:814-823.
10. Griffith DE, et al. ATS 2018, abstract A5915.

ACKNOWLEDGMENTS

- We thank the patients and their families for their support and participation, and the study investigators, study coordinators, and support staff across all sites.
- Editorial assistance was provided by Richard Boehme, PhD, of MediTech Media, Ltd, and funded by Insmed Incorporated.
- Services for MAC culture, identification, and susceptibility testing were provided by the University of Texas Health Science Center, Tyler, Texas; Radboud University Medical Center, Nijmegen, the Netherlands; and the Queensland Mycobacterium Reference Laboratory, Herston, Queensland, Australia.

DISCLOSURES

- KL Winthrop has received grant support and consulting fees from Insmed Incorporated.
- DE Griffith has received consulting fees from Insmed Incorporated.
- Financial support for this study was provided by Insmed Incorporated.

Copies of this poster obtained through the QR Code are for personal use only and may not be reproduced without permission from the author of this poster.

Poster presented at the ATS 2018, 18-23 May 2018, San Diego, CA.