Inhibition of Neutrophil Serine Protease Activation with the Reversible DPP1 Inhibitor AZD7986

J. Root[1], J. Bernström[1], S. Clifton[2], G. Dahl[1], K. Doyle[2], P. Gardiner[1], P. Johannesson[1], H. Käck[1], H Lönn[1], J. Shearer[2], C. Wikell[1], K. Stenvall[1]

AstraZeneca R&D, Gothenburg, Sweden[1]; Charles River Discovery, Cambridge, UK[2]

P1328

Abstract

Neutrophils and their destructive, pro-inflammatory cargo of proteases are implicated as key players of several chronic respiratory disorders[1]. The DPP1 inhibitor AZD7986 is a reversible, potent, inhibitor of the protease DPP1. The inhibitory potency of AZD7986 correlates well across multiple species. We show significant reductions of active neutrophil serine proteases (NSPs) *in vitro* using neutrophil precursor cells, and *in vivo* using naïve rats. The reduction of activated NSPs within neutrophils will decrease the inflammation and tissue destruction caused by active neutrophilic disease.

Introduction

- Neutrophils, and the serine proteases stored within their azurophilic granules, are implicated in the pathology of obstructive lung disorders such as bronchiectasis, CF and COPD. Further increases in NSP activity have been reported during exacerbations[2].
- NSPs are activated by the lysosomal cysteine protease dipeptidyl peptidase 1 (DPP1, also known as cathepsin C) during neutrophil maturation in the bone marrow.
- DPP1 is expressed in both haematopoietic and non-haematopoietic cells.
- Loss-of-function mutations of DPP1 in humans result in the loss of active NSPs, and result in disorders such as Papillon-Lefèvre syndrome (PLS)[3].
- NSPs can induce and potentiate inflammation[4].
- NSPs decrease mucociliary clearance by promoting mucus production & release[5].
- NSPs are stored within granules at ~mM levels[6], release of which (illustrated in Figure 1) in disease where a protease-antiprotease imbalance exists can act unopposed further from the origin of release[7]. This approach of inhibition of activation could be more effective than inhibition of activity.

Methods

In Vitro Studies

- Binding kinetics were assessed using a Biacore T200 (GE Healthcare) Surface Plasmon Resonance direct binding assay), with immobilised human DPP1.
- Potency of DPP1 inhibition was assayed using isolated recombinant DPP1 enzyme and the monocytic U937 cell line. Fluorescent synthetic dipeptide substrates were used to measure DPP1 activity.
- The potencies of AZD7986 at DPP1 of non-human species were obtained using recombinant isolated enzyme.
- Human CD34+ neutrophil precursor cells were differentiated using SCF, IL-3, G-CSF and different concentrations of AZD7986 for 7 days. Cell lystates were then measured using synthetic peptide substrates and concentration response curves fitted to obtain potency values.

In Vivo Studies

Naïve male Sprague-Dawley rats were dosed *p.o.* with AZD7986 for 8 days, after which bone marrow cells were isolated and NSP activities assessed using peptide substrates.

Results

- AZD7986 binds reversibly to DPP1, with a residence time of 3 min (Figure 2).
- Concentration dependent inhibition of DPP1 was observed in both the enzyme and U937 cell-based assays, with similar mean pIC_{50} values of 8.35±0.08 and 8.37±0.04 (mean±SEM, n=10 & 14) respectively.
- Good species crossover was seen with AZD7986 for inhibiting mouse, rat, rabbit and dog DPP1: 2.7, 2.4, 1.7 and 2-fold less than that for human DPP1 respectively (n=3-8)
- All 3 NSP activities studied were concentration dependently decreased to negligible levels in neutrophil precursor cells with AZD7986, with pIC_{50} values of 7.21±0.14, 6.68±0.11 and 6.94±0.18 respectively (mean±SD, n=4, Figure 3).
- Rat bone marrow cell lysate NSP activities were significantly inhibited by AZD7986 (Figure 4).

Figure 1. Neutrophil elastase released by human neutrophils purified from peripheral blood.
Neutrophils are stained blue with DAPI, elastase is stained red with mouse anti-human elastase mAb; James Root & Holger Schluter, AstraZeneca.

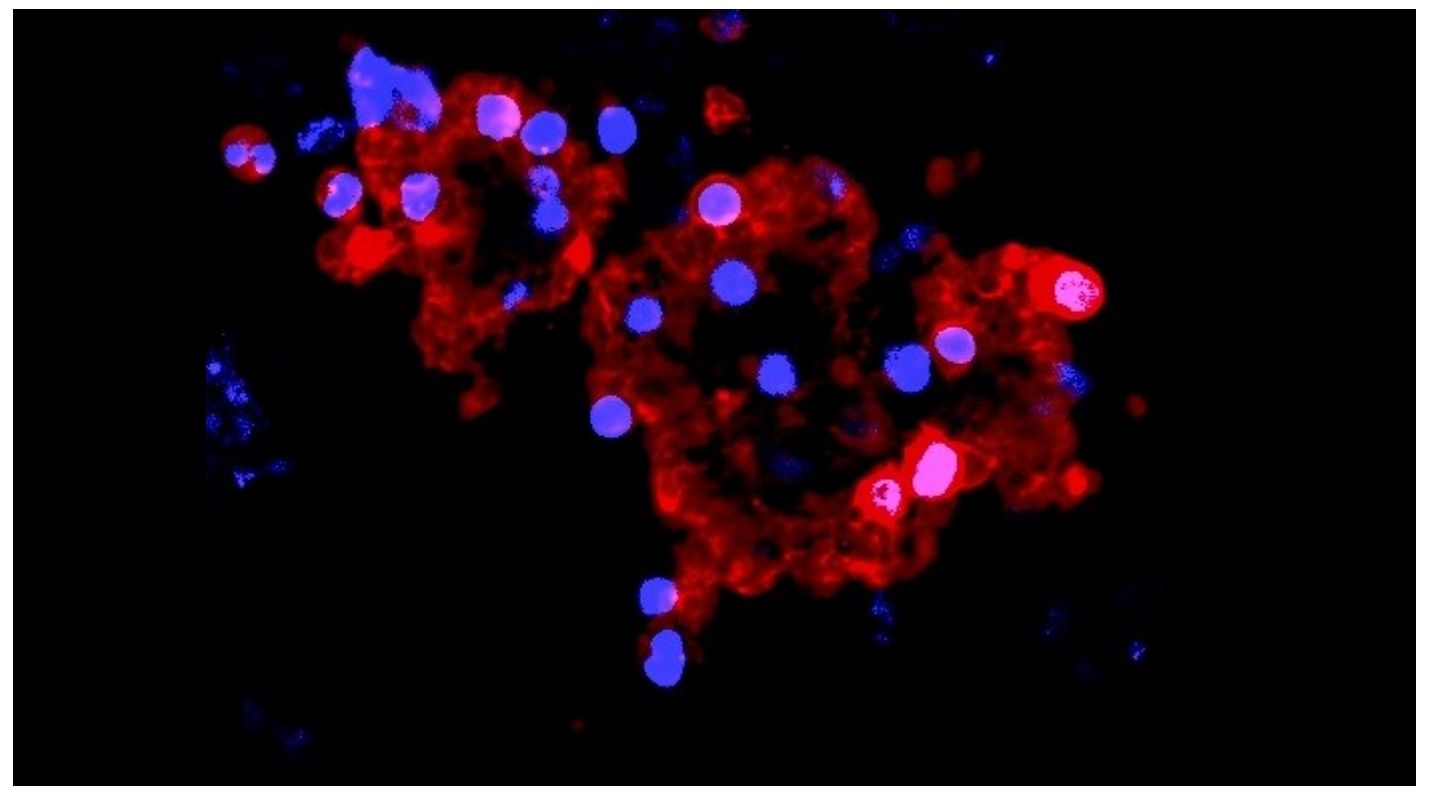

Figure 2. Compound sensorgram showing reversibility of AZD7986 binding to human DPP1

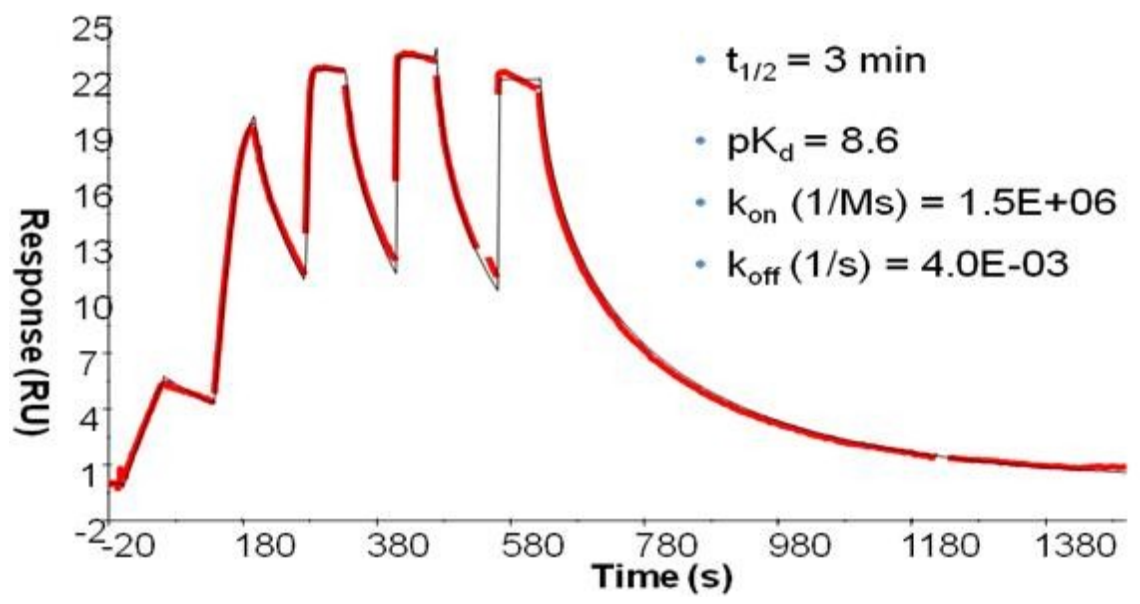
Response (RU)
Time (s)
-2
1
4
7
10
13
16
19
22
25
-20
180
380
580
780
980
1180
1380
$t_{1/2}$ = 3 min
pK_d = 8.6
k_{on} (1/Ms) = 1.5E+06
k_{off} (1/s) = 4.0E-03

Figure 3. DPP1 inhibition using AZD7986 in human neutrophil precursor cells results in decreased activated NSPs
Data are mean±SD, n=4.

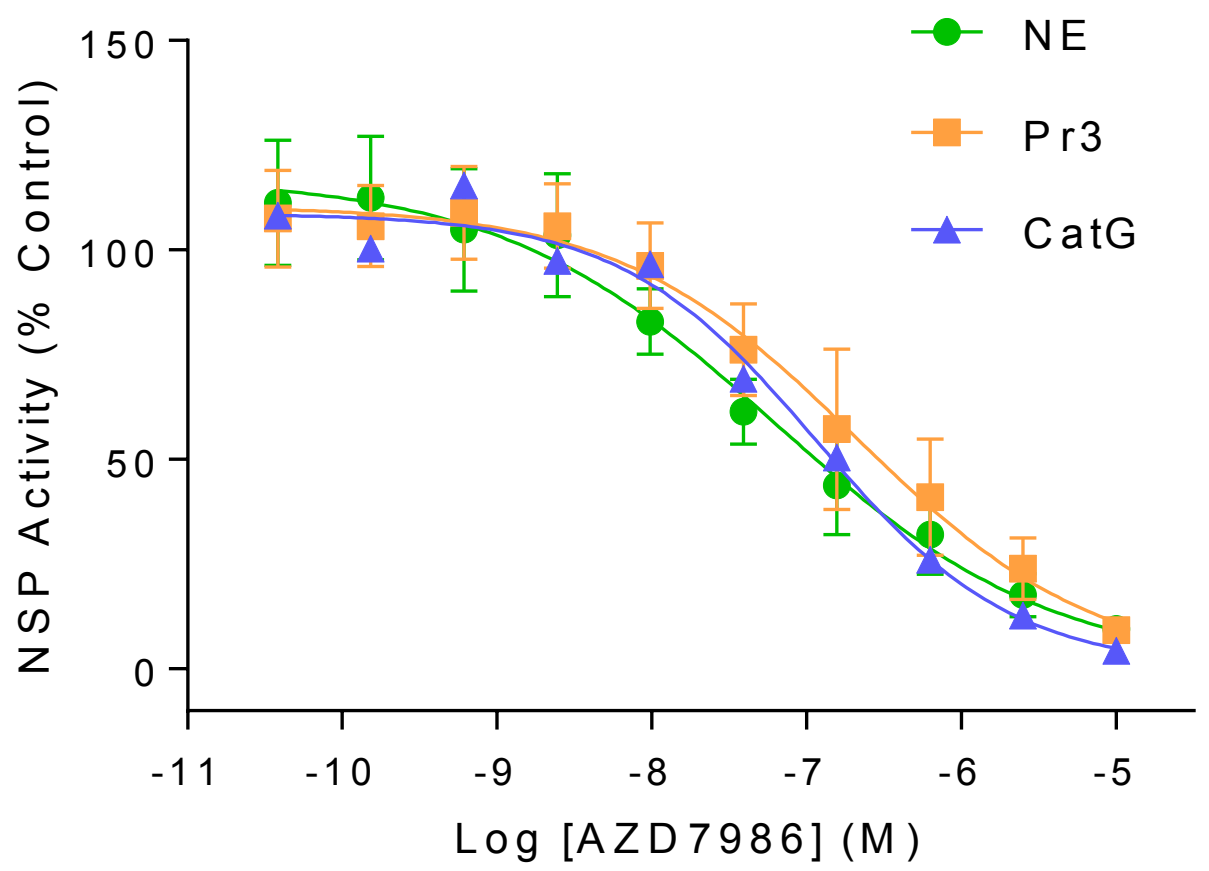
NSP Activity (% Control)
0
50
100
150
-11
-10
-9
-8
-7
-6
-5
Log [AZD7986] (M)
NE
Pr3
CatG

Conclusions

- AZD7986 is a potent and reversible small-molecule inhibitor of isolated and cellular human DPP1.
- AZD7986 potency shows good species crossover, which increases confidence in the translation of pre-clinical data to patients.
- Both *in vitro* and *in vivo,* AZD7986 treatment results in significant reduction in activated NSPs.
- AZD7986 could be a more effective approach to redressing protease-antiprotease imblances compared to classical NSP inhibitors, and may benefit patients with active neutrophilic disorders, such as COPD.

Figure 4. AZD7986 inhibits activation of NSPs in bone marrow cell lysates when administered for 8 days to naïve rats.
Data are expressed as a percentage of the control group, mean±SEM, n=10.

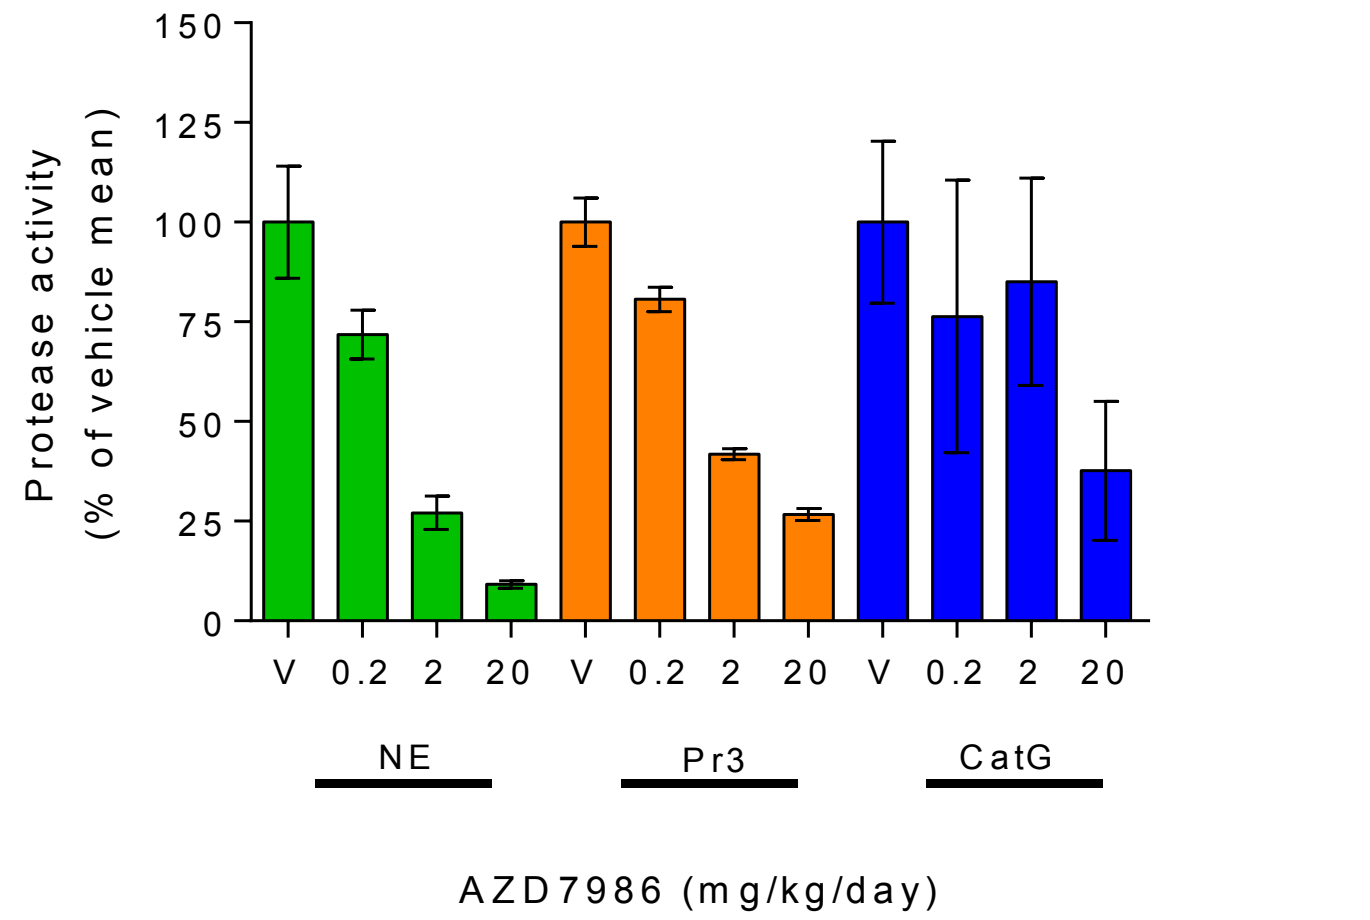
Protease activity
(% of vehicle mean)
0
25
50
75
100
125
150
V
0.2
2
20
V
0.2
2
20
V
0.2
2
20
NE
Pr3
CatG
AZD7986 (mg/kg/day)

References

1. Barnes, PJ., Clin Chest Med. 2014 Mar;35(1):71-86
2. Sinden, NJ, Eur Respir J. 2013 May;41(5):1042-50
3. Pham, CT, J Immunol. 2004 Dec 15;173(12):7277-81
4. Kessenbrock, K., J Mol Med (Berl). 2011 Jan;89(1):23-8
5. Park, JA,. Am J Pathol. 2005 Sep;167(3):651-61
6. Liou and Campbell, Biochemistry. 1995 Dec 12;34(49):16171-7
7. Campbell, EJ., J Clin Invest. 1999 Aug 1; 104(3): 337–344

Acknowledgements

Multiple staff at AstraZeneca Gothenburg and Charles River who contributed to the experimental work; Holger Schluter who help produce the confocal fluorescence microscopy image (Figure 1).

Supported by AstraZeneca IMED Biotech Unit

Presented at the 2016 San Fransisco ATS, James Root